Noncovalent electrospray ionization mass spectrometry: a powerful tool in drug discovery

Kurt Benkestock,[1,2] Per-Olof Edlund [1] and Johan Roeraade [2]

[1]Biovitrum AB, SE-112 76, Stockholm, Sweden and [2]Royal Institute of Technology, SE-100 44, Stockholm, Sweden. E-mail: kurt.benkestock@biovitrum.com

Introduction

The vast number of new drug candidates produced by high throughput chemistry (HTC) requires fast and efficient methods both for characterization of the individual compounds and their affinity to target molecules. One way to accelerate drug discovery and thus decrease the time for the overall drug development process is to perform drug screening at an early stage of the process. A prerequisite for such an approach is the availability of sensitive analytical methods.

The advantages of using electrospray mass spectrometry (ESI-MS) for non-covalent binding studies are the high sensitivity, the speed of analysis, the ability to obtain stoichiometric information, and the mass measurement accuracy. Furthermore, there is the possibility to determine dissociation constants (K_d) of non-covalent weakly bound complexes.

Noncovalent electrospray ionization mass spectrometry (ESI-MS) has succesfully been introduced at Biovitrum as a powerful routine based screening tool in drug research.[1] The principle is simple, the analyses is fast and consumes low amount of target proten. Protein and ligand are mixed prior to injection into the MS-instrument. Binding is confirmed by detecting the noncovalent protein-ligand complex in the gas-phase.

$$P + L \overset{K_d}{\rightleftharpoons} PL$$

Ligand screening

Depending on the nature of the samples, analysis is performed with or without a microdialysis device. Many proteins easily form metal adduct ions which impair their analysis by MS. The microdialysis device eliminates unwanted adducts with enhance sensitivity and interpretation of spectra as shown in Figure 2.

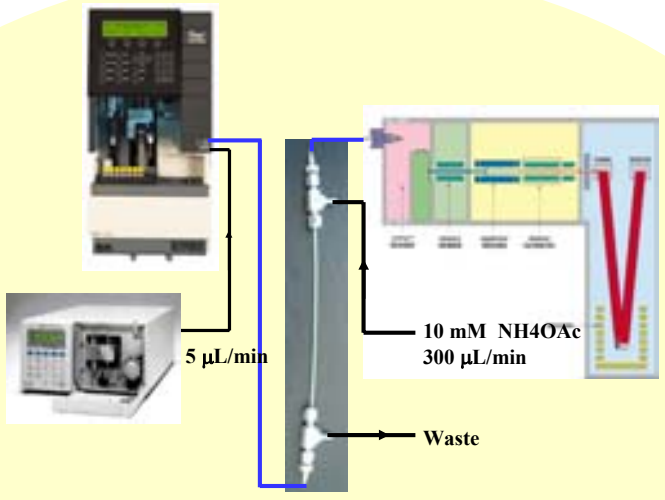
5 µL/min
10 mM NH4OAc
300 µL/min
Waste

Fig. 1. Ligand screening instrument set-up with microdialysis device

- Sample capacity approximate 70 samples/day.
- Protein consumption in the range of 500 picomoles/sample.

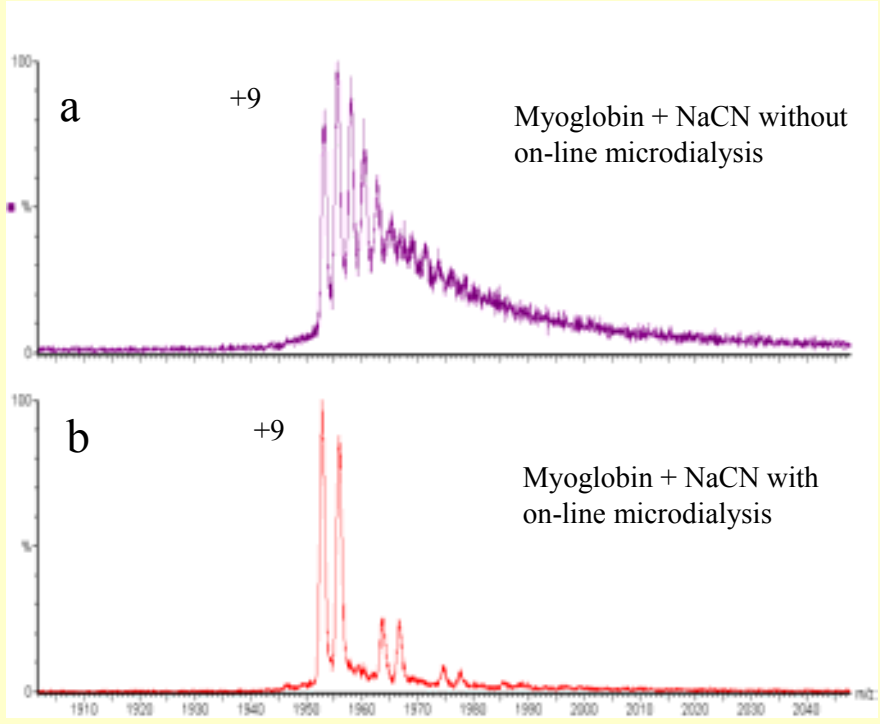
a
+9
Myoglobin + NaCN without on-line microdialysis
b
+9
Myoglobin + NaCN with on-line microdialysis

Fig. 2. ESI mass spectra of horse heart myoglobin (27 µM) with NaCN solution added to give a final concentration of 0.05 µM. (a) Without the on-line microdialysis, and (b) with on-line microdialysis.

Chip-based ligand screening

The core of the system consisted of a chip-based platform for automated sample delivery from a 96-well plate (Advion Bioscience) and subsequent analysis by nano-ESI based on non-covalent interactions.[2]

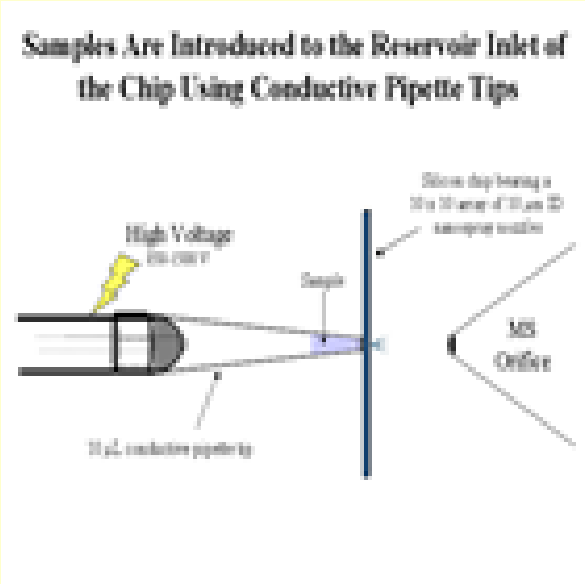
Samples Are Introduced to the Reservoir Inlet of the Chip Using Conductive Pipette Tips
High Voltage
Sample
MS
Orifice

- Sample capacity approximate 430 samples/day.
- Protein consumption in the range of 100 picomoles/sample.

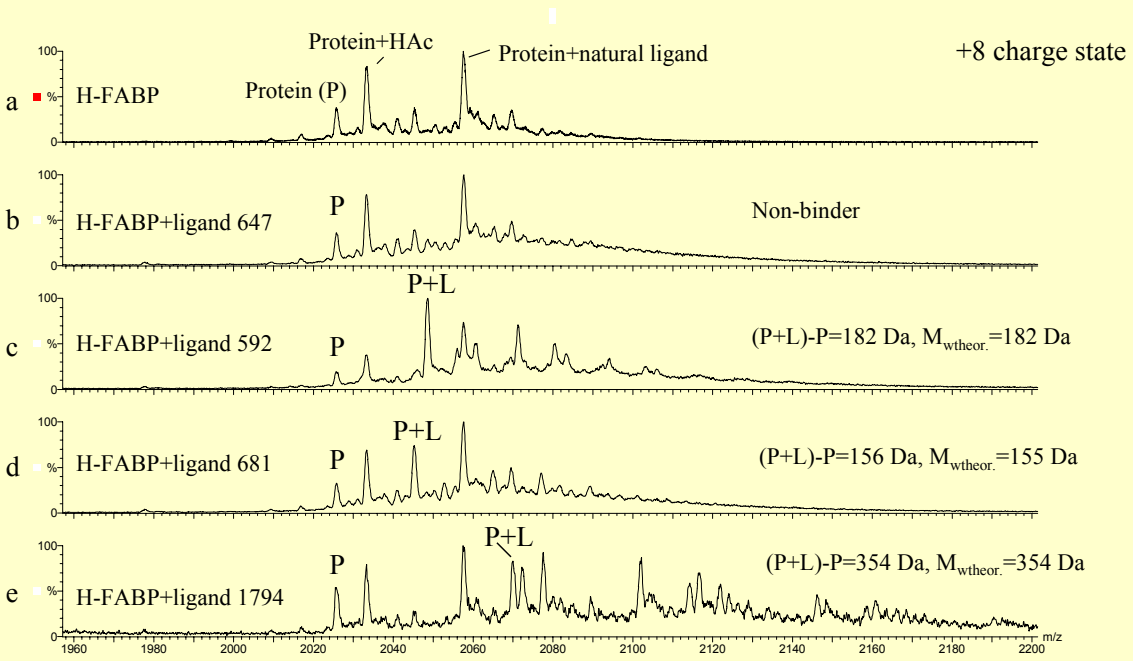
+8 charge state
a
H-FABP
Protein (P)
Protein+HAc
Protein+natural ligand
b
H-FABP+ligand 647
P
Non-binder
c
H-FABP+ligand 592
P
P+L
(P+L)-P=182 Da, Mwtheor.=182 Da
d
H-FABP+ligand 681
P
P+L
(P+L)-P=156 Da, Mwtheor.=155 Da
e
H-FABP+ligand 1794
P
P+L
(P+L)-P=354 Da, Mwtheor.=354 Da
1960
1980
2000
2020
2040
2060
2080
2100
2120
2140
2160
2180
2200
m/z

Fig. 3. Spectrum a) shows the human fatty acid binding, H-FABP (heart) target protein. Spectra d-e) shows H-FABP mixed with potential ligands using the chip-based system. The natural ligand was partly displaced by the different binders.

Determination of dissociation constants

We are using a novel approach for the calculation of the dissociation constants of weakly bound complexes. The method is based on competitive binding using a dose-response titration followed by calculation of K_d from a non-linear regression curve.

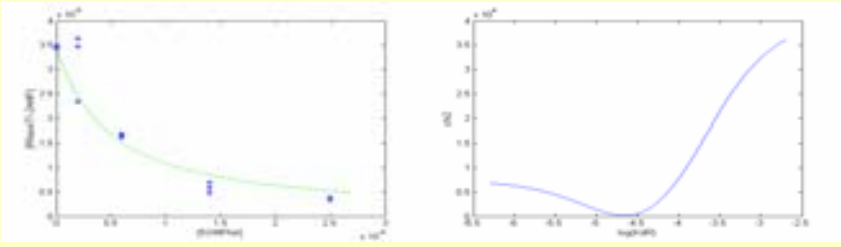

Fig. 4. Determination of dissociation constant for a protein-ligand complex. The best fitted curve was found for $K_{d,}$=21.8µM. b) χ^2 as a function of $K_{d,}$, minimum at $\log K_{d,}$=-4.66

For more details see poster number: ThC-138 by A. Tjernberg *et al.*

Conclusions

- Noncovalent ESI/MS has had great impact in many research projects, especially in projects where other screening methods were difficult to develop.
- Automatic nano-ESI/MS has great potential to serve as a complementary screening method to conventional HTS. Alternatively, it could be used as a first screening method in an early phase of drug development programs, where only small amounts of purified protein are available.
- Determinations of dissociation constants of protein-ligand complexes are done on routine basis using a novel approach.

Interested to read more?

1. Benkestock *et al*, Rapid Communication in Mass Spectrometry 2002; 16: 2054 2059
2. Benkestock *et al*, Journal of Biomolecular Screening 2003; 8(3): 247 256